PRELIMINARY ASSOCIATED CHARACTERISTICS OF THE TOP 10% OF GLP-1 SPENDERS IN A SELF-INSURED HEALTH PLAN

Authors: Huyen Vu, Pharm.D.[1]; Shellie Keast, Pharm.D., Ph.D.[1]; Wenyi Qiu, M.S.[1]; Charlene Dohanh, Pharm.D.[1]; Jennifer Hendricks, Pharm.D., APh, BCACP, CDCES, FCPhA[1]
Affiliations: 1. MedImpact Healthcare Systems, Inc, San Diego, CA, USA

INTRODUCTION

- Obesity affects approximately 42% of adults in the US[1] while Type 2 Diabetes affects around 10% of the adult population.[2]
- Up to 53% of diabetes development can be attributed to obesity.[3]
- The utilization of GLP-1 therapies has increased substantially in recent years, resulting in a sharp rise in medication costs for health plans.[4]
- Identifying characteristics of the top spenders on GLP-1 drugs may support strategic planning for cost control within this drug class.

OBJECTIVE

To determine the characteristics associated with the top 10% of GLP-1 spenders in a large, self-insured health plan.

METHODOLOGY

- A self-insured commercial health plan covering over 18,000 members, primarily in Western states, was analyzed.
- The study included all GLP-1 pharmacy claims from 01/01/2024 to 12/31/2024.
- Top spenders were defined as the top 10% of members based on total plan-paid costs for GLP-1 therapies.
- Statistical comparisons between top and non-top spenders were conducted across age, gender, GLP-1 utilization patterns, diagnosis (based on ICD-10 codes recorded in MedImpact Pharmacy Claim data), comorbidities (hypertension and hyperlipidemia), and appropriateness of use.
- Logistic regression was performed on selected variables to identify predictors of high spending.

Figure 1. Patient Identification Flow Chart

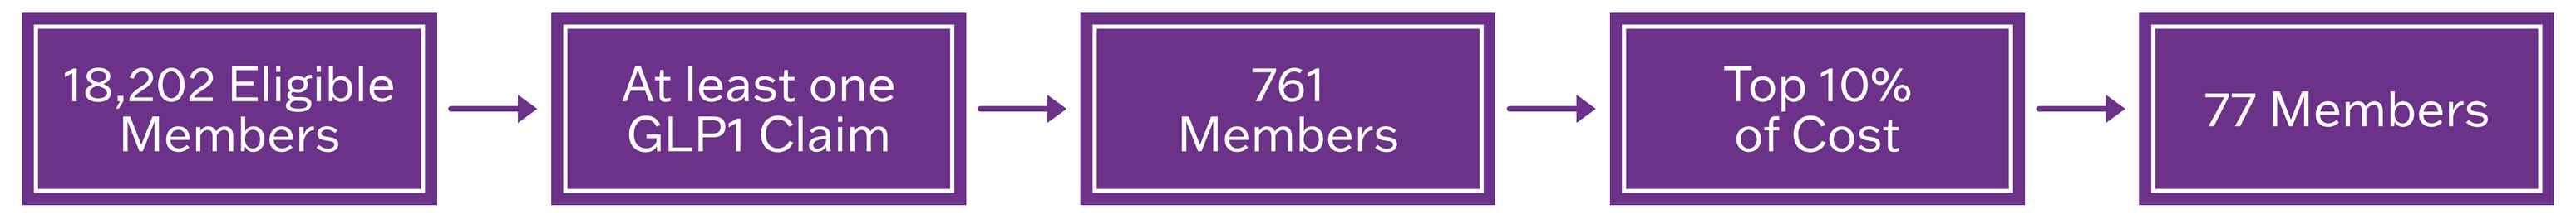
18,202 Eligible Members
At least one GLP1 Claim
761 Members
Top 10% of Cost
77 Members

RESULTS

Figure 2. Percent of Overall Members on GLP-1s by Diagnosis

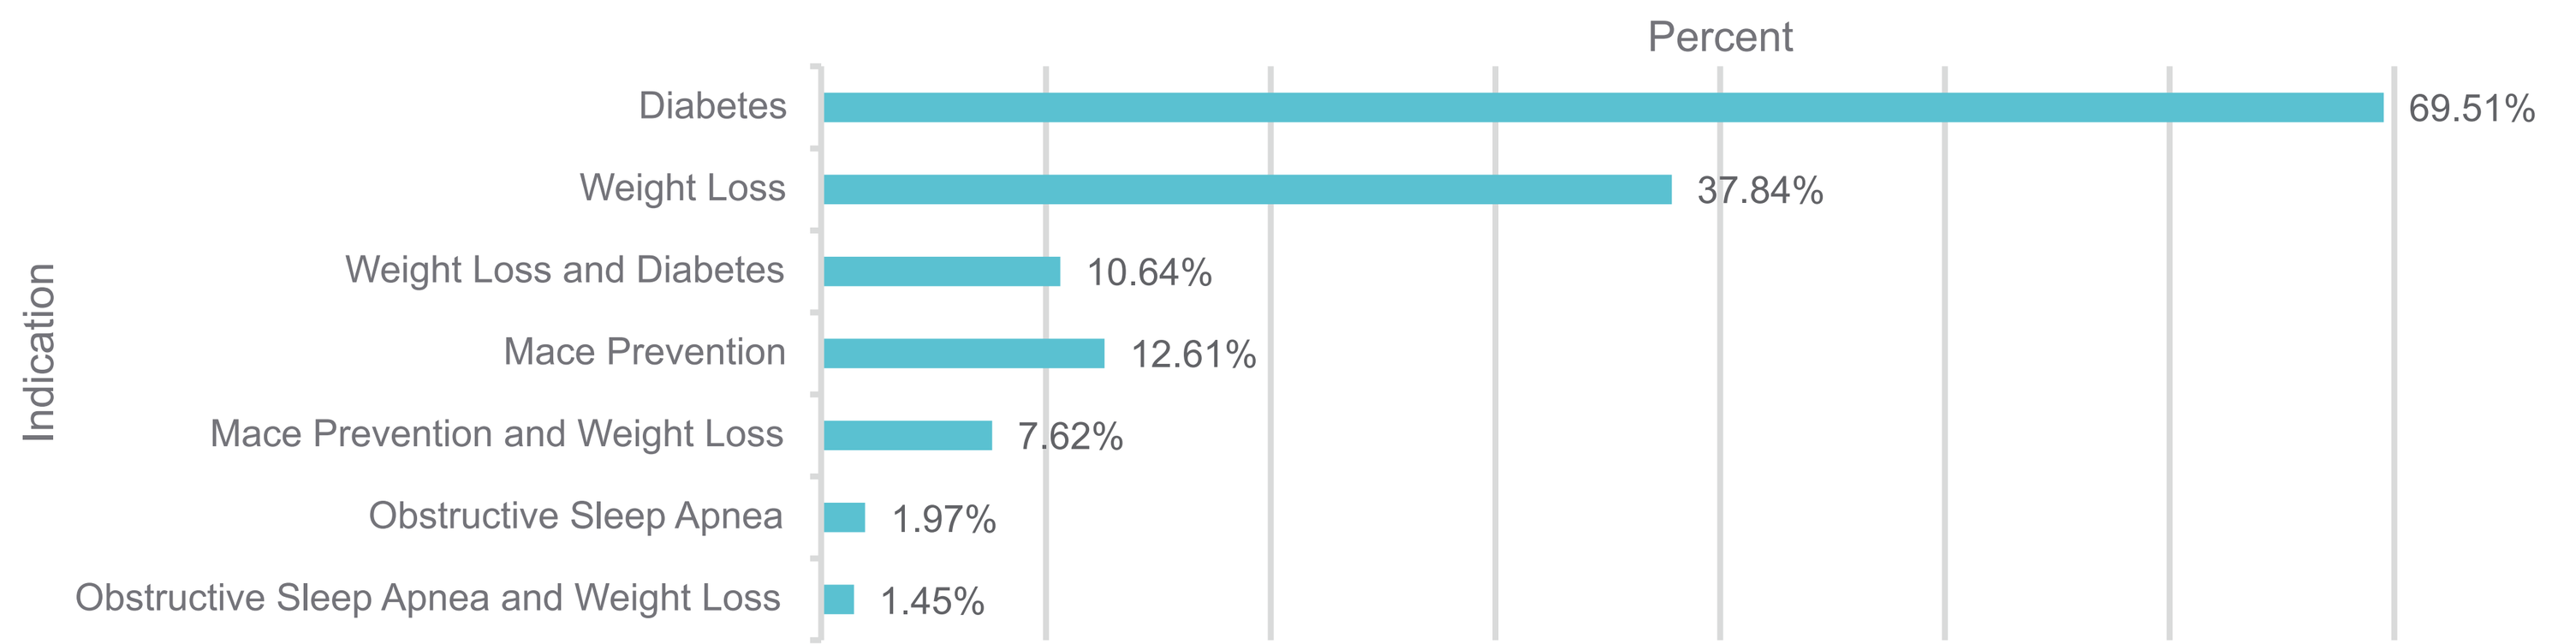
Percent
Indication
Diabetes
69.51%
Weight Loss
37.84%
Weight Loss and Diabetes
10.64%
Mace Prevention
12.61%
Mace Prevention and Weight Loss
7.62%
Obstructive Sleep Apnea
1.97%
Obstructive Sleep Apnea and Weight Loss
1.45%

Figure 3. Top 10% of GLP-1 Spenders

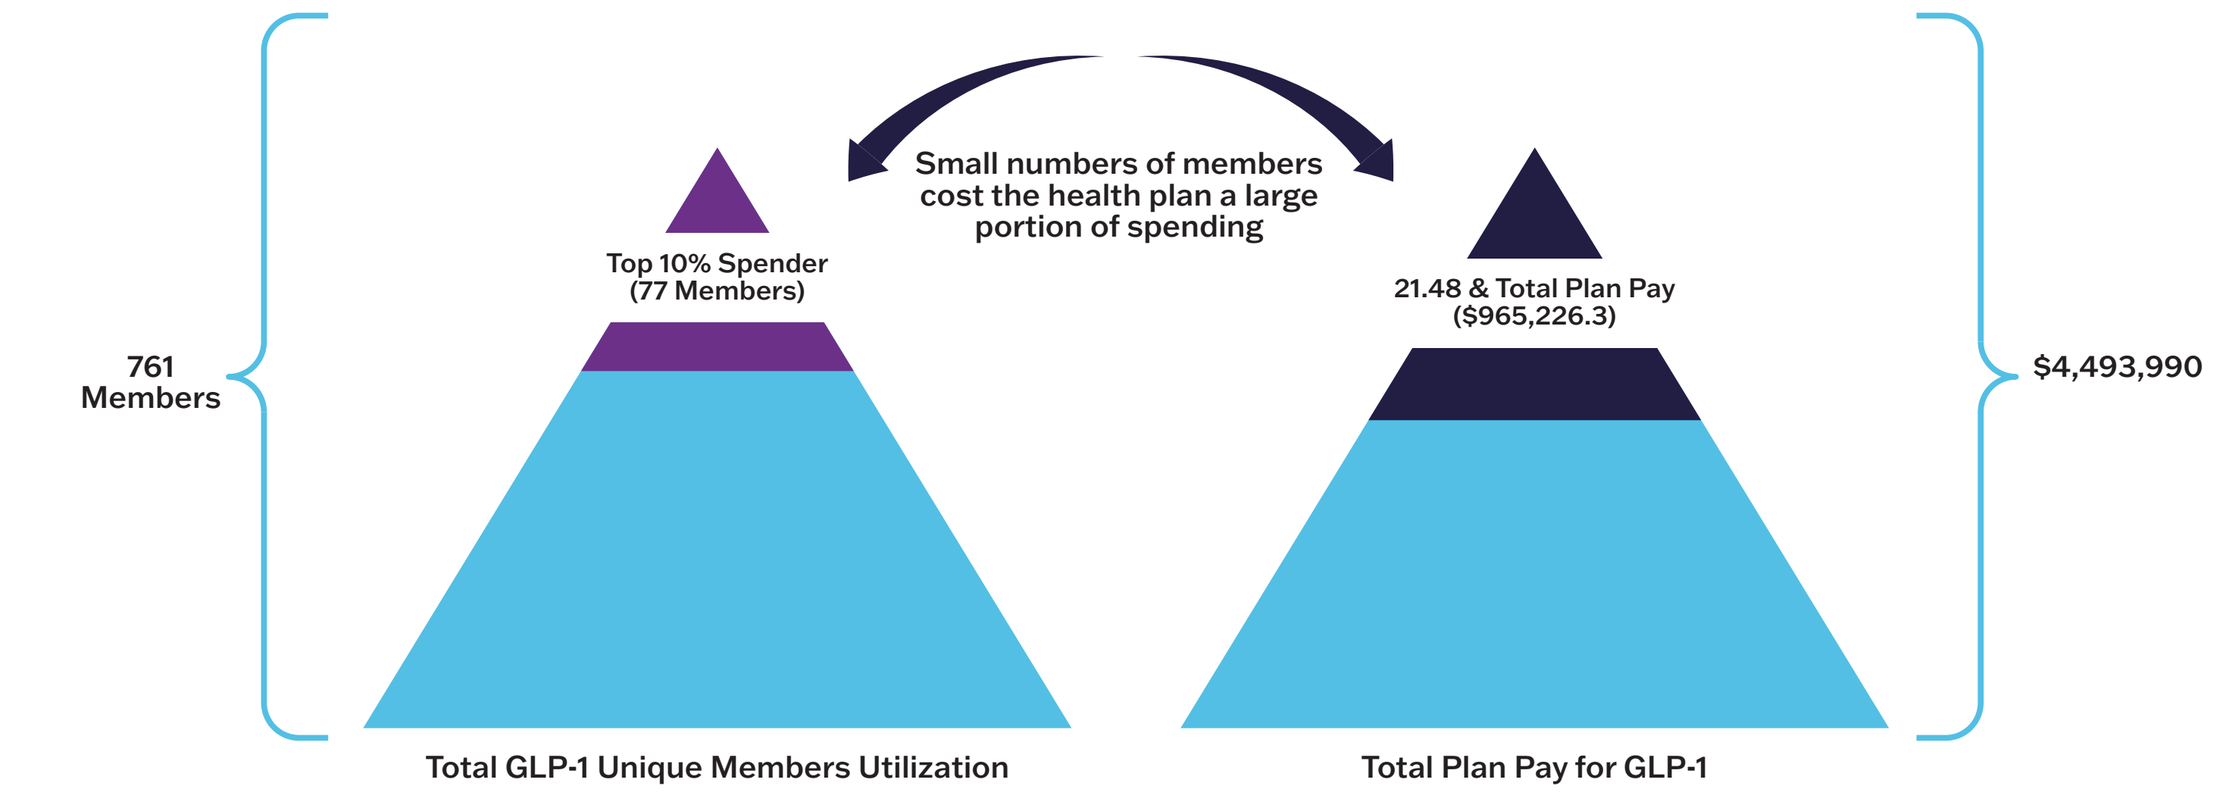
Small numbers of members cost the health plan a large portion of spending
Top 10% Spender (77 Members)
761 Members
21.48 & Total Plan Pay ($965,226.3)
$4,493,990
Total GLP-1 Unique Members Utilization
Total Plan Pay for GLP-1

Table 1. Overall Usage and Cost of GLP-1 Agonists

Description	Values
Total Numbers of Unique Member on GLP-1	761
Average Age per Unique Member on GLP-1	51.68
Average Total Ingredient Cost per Unique Member on GLP-1	$ 6,124.60
Average Numbers of GLP-1 Claims per Unique Member on GLP-1	4.96
Average Numbers of Days Supply per Unique Member on GLP-1	175.56
Average Numbers of Unique GLP-1 Therapies per Utilized Member	1.1
Average Numbers of Diagnosis per Unique Member on GLP-1*	1.28
Average Numbers of Approved Indications Diagnosis per Unique Member on GLP-1	1.22
Average Numbers of Comorbidities per Unique Member on GLP-1	1.13

Table 2. Overall Usage by Demographics and Diagnoses

Variables	Subcategories	Number of Members (N (%))
Age Group	≤ 50 years old	316 (41.52 %)
	> 50 years old	445 (58.48 %)
Gender	Female	503 (66.1 %)
	Male	258 (33.9 %)
Region	West	754 (99.08 %)
	Mid West	3 (0.39 %)
	South	4 (0.53 %)
Numbers of Unique GLP-1 Brand Drugs	1	695 (91.33 %)
	2	65 (8.54 %)
	3	1 (0.13 %)
Numbers of Diagnosis	1	564 (74.11 %)
	2	179 (23.52 %)
	3	17 (2.23 %)
	4	1 (0.13 %)
Comorbidities	Hyperlipidemia	391 (51.38 %)
	Hypertension	472 (62.02 %)
Numbers of Comorbidities	0	209 (27.46 %)
	1	241 (31.67 %)
	2	311 (40.87 %)

Figure 4. Overall Usage Stratified by Brand Name and GLP-1 Type

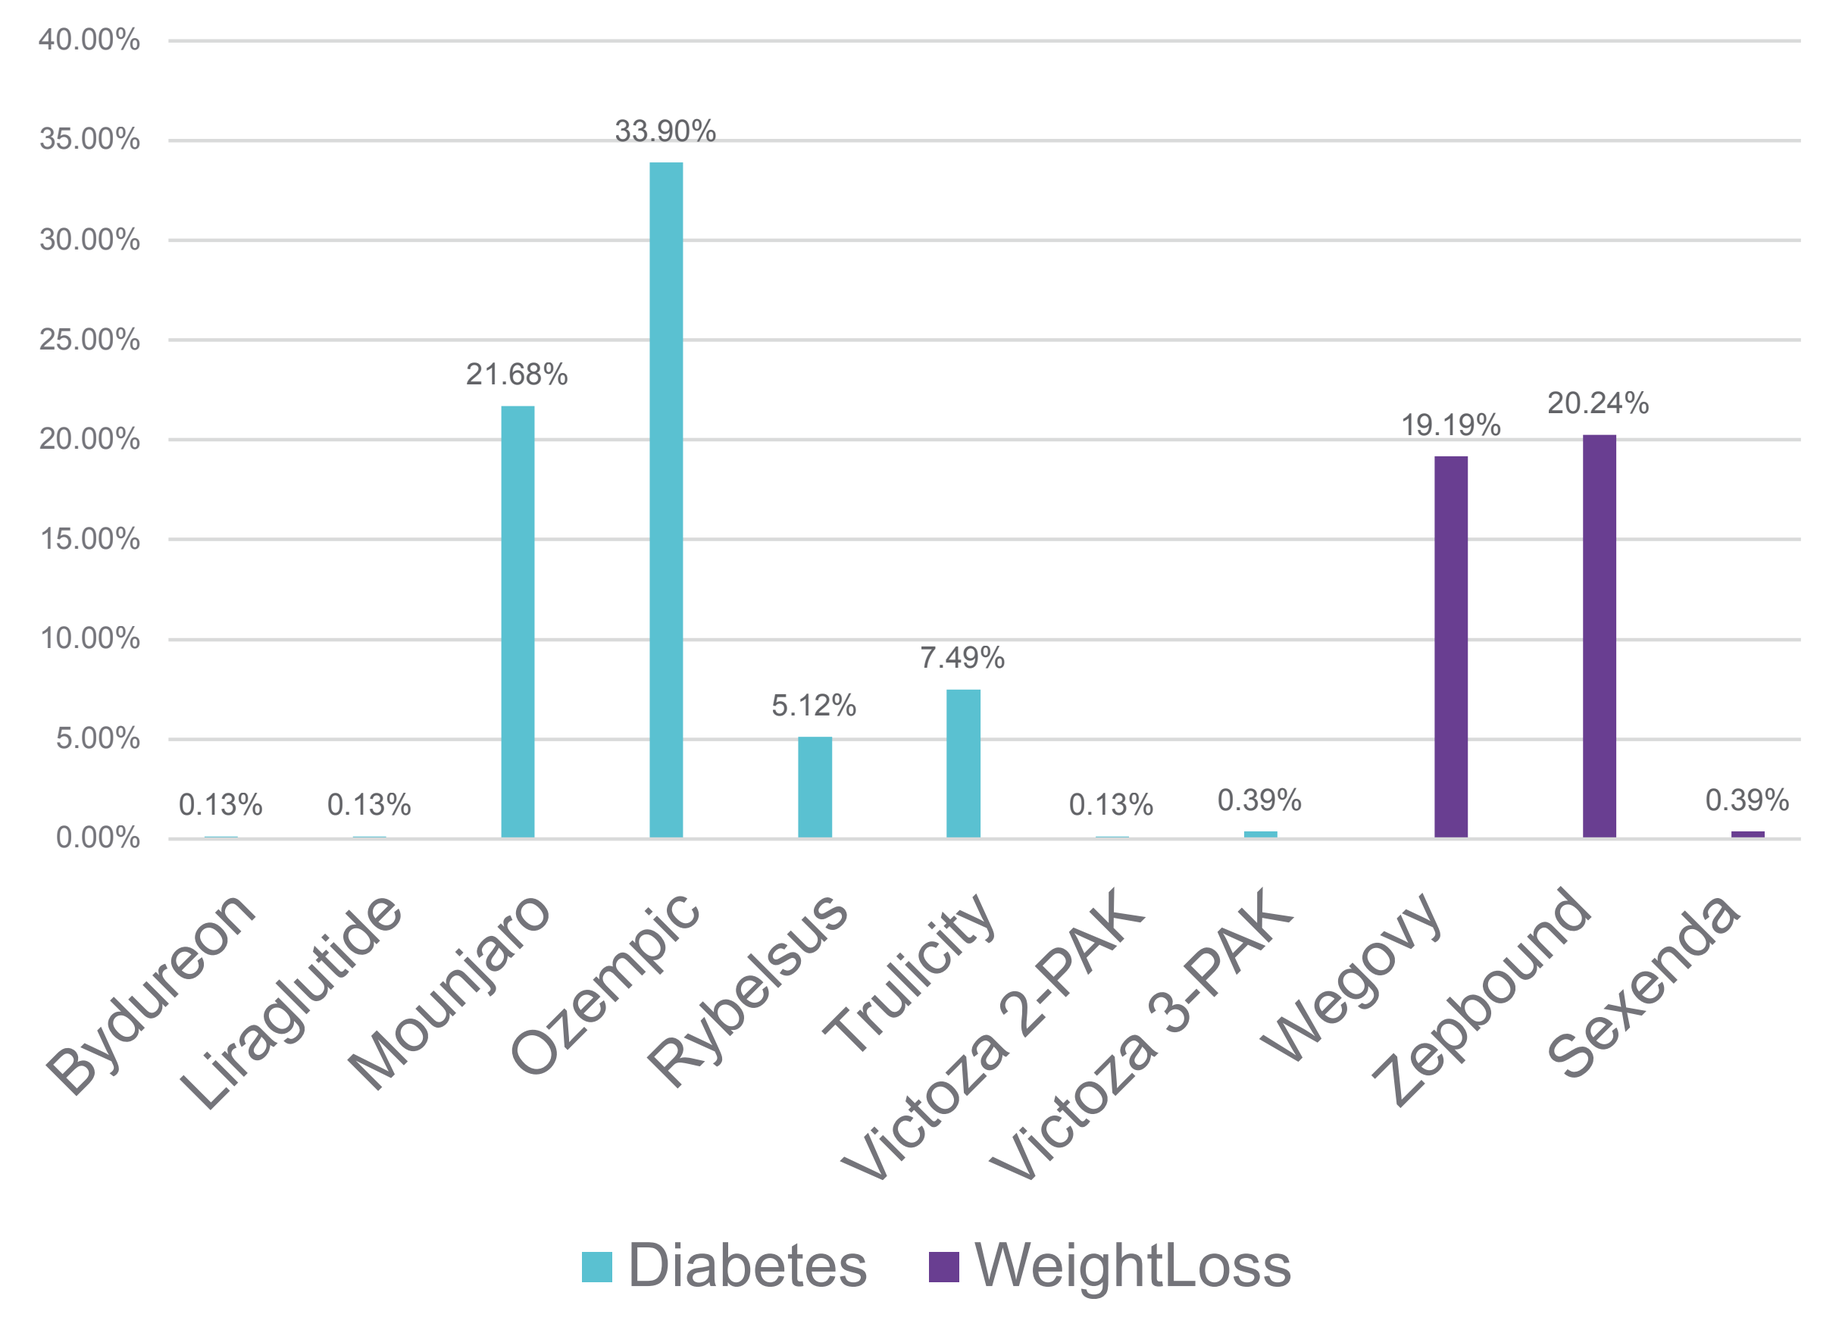
40.00%
35.00%
30.00%
25.00%
20.00%
15.00%
10.00%
5.00%
0.00%
0.13%
0.13%
21.68%
33.90%
5.12%
7.49%
0.13%
0.39%
19.19%
20.24%
0.39%
Bydureon
Liraglutide
Mounjaro
Ozempic
Rybelsus
Trulicity
Victoza 2-PAK
Victoza 3-PAK
Wegovy
Zepbound
Sexenda
Diabetes
WeightLoss

Table 3. Comparison between Top 10% Spenders and Non-Top 10% Spenders

Characteristic		Top 10 % Spenders(N=77)	Non – Top Spenders(N=684)	P value
Age (Average (SD))		56.34 (8.60)	51.16 (11.36)	0.0001
Age Group (N (%))				
	≤ 50 years old	17 (22.08 %)	299 (43.71 %)	0.0003
	> 50 years old	60 (77.92 %)	385 (56.29 %)	
Gender (N (%))				
	Female	46 (59.74 %)	457 (66.81 %)	0.2139
	Male	31 (40.26 %)	227 (33.19 %)	
Numbers of Claims (Mean (SD))		9.08 (2.99)	4.50 (2.92)	<0.0001
Numbers of Days Supply (Mean (SD))		355.8 (49.67)	155.3 (96.19)	<0.0001
Total Ingredient Cost (Mean (SD))		$ 12,909.10 ($ 1,480.70)	$ 5,360.90 ($ 3,169.70)	<0.0001
Total Unique GLP-1 (Mean (SD))		1.14 (0.35)	1.08 (0.28)	0.0783
Number of Diagnosis (Mean (SD))		1.25 (0.52)	1.29 (0.51)	0.498
Number of Comorbidities (Mean (SD))		1.39 (0.76)	1.11 (0.82)	0.0037
Comorbidities Numbers Categories (N (%))				0.0128
	0	13 (16.88 %)	196 (28.65 %)	
	1	21 (27.27 %)	220 (32.16 %)	
	2	43 (55.84 %)	268 (39.18 %)	
Hypertension		68.83%	61.26%	0.1942
Hyperlipidemia		70.13%	49.27%	0.0005
Weight Loss		31.17%	38.60%	0.2026
Diabetes		76.62%	68.71%	0.1529
Weight Loss and Diabetes		10.39%	10.67%	0.9392
MACE Prevention		1.31%	12.57%	0.9174
MACE Prevention and Weight Loss		9.09%	7.46%	0.6083
Obstructive Sleep Apnea		1.30%	2.05%	0.6544
Obstructive Sleep Apnea and Weight Loss		1.30%	1.46%	0.9094

Table 4. Logistic Regression Results

Variables		Odd Ratio	P value
Age Group	≤ 50 years old	0.426	0.0065
	> 50 years old	Ref	
MOUNJARO	Yes	2.687	0.001
	No	Ref	
WEGOVY	Yes	4.403	<0.0001
	No	Ref	
ZEPBOUND	Yes	0.116	0.037
	No	Ref	
Hyperlipidemia	Yes	2.136	0.1166
	No	Ref	
Comorbidities Numbers Categories	0	Ref	0.972
	1	0.901	
	2	0.877	

DISCUSSION

For this population:

- Utilizing Wegovy and utilizing Mounjaro are strong positive predictors of being in the top 10% spender
 - Being on Wegovy is 4.4 times more likely to be in the top 10% spender
 - Being on Mounjaro is 2.687 times more likely to be in the top 10% spender
- Younger GLP-1 utilizers are less likely to be in the top 10 % spenders
- Utilizing Zepbound has a negative relationship with being in the top 10% spenders
- Likely to be in the top 10% spenders with an OR of 0.223
- Numbers of comorbidity burden and hyperlipidemia are not significant factors that influence the model outcomes

Limitations

- Results of the analysis are from a single population and may not be generalizable to the larger population
- Analysis utilized pharmacy claims data with some assumptions of use of GLP-1 products for diabetes and weight loss
- Cost data is not inclusive of rebates
- Specific product usage may be biased due to plan formulary requirements

CONCLUSION

This study identified key characteristics of the top 10% of GLP-1 spenders: older age, use of Wegovy and Mounjaro, and lack of Zepbound use. Targeted educational and strategic initiatives may help control costs and improve outcomes. These findings are limited to one large, self-insured commercial plan; broader studies are needed to generalize the results.

REFERENCES

1. New CDC Data Show Adult Obesity Prevalence Remains High. 2024, Center for Disease Control and Prevention.
2. Type 2 Diabetes. May 15,2024 [cited 2025 August 14, 2025]; Available from: https://www.cdc.gov/diabetes/about/about-type-2-diabetes.html.
3. Cameron, N.A., et al., Quantifying the Sex-Race/Ethnicity-Specific Burden of Obesity on Incident Diabetes Mellitus in the United States, 2001 to 2016: MESA and NHANES. Journal of the American Heart Association, 2021. 10(4): p. e018799.
4. Mahase, E., GLP-1 agonists: US sees 700% increase over four years in number of patients without diabetes starting treatment. BMJ, 2024. 386: p. q1645.